TLR4 Agonist GLA Modifies Th1/2 Cytokine Profiles in PBMC from Patients with Pollen Allergy

Hailing Lu[1], Richard Roque[2], Jan ter Meulen[1], Christopher H. Clegg[2]
[1]Immune Design, Seattle, WA; [2]TRIA Biosciences, Seattle, WA

BACKGROUND

- **Allergy is characterized by a Th2-biased immune response.**
- **Allergen-specific Immunotherapy (SIT) is the curative treatment for allergy and involves subcutaneous injection of increasing doses of allergen extract (SCIT) or sublingual administration of allergen extract (SLIT).**
- **The focus of SIT is to shift Th2 bias to Th1.**
- **GLA is a novel TLR4 agonist at the core of the GLAAS™ (GLA Adjuvant System) discovery platform that may hold potential for SIT due to its ability to activate DC and polarize Th1 immune responses.**
 - GLA induces the expression of co-stimulatory molecules, CD80, CD86, and CD40 on DC (Ref 1).
 - GLA induces IL-12, TNFα, and other pro-inflammatory cytokines (Ref 1 and 2).
 - GLA is a clinical-stage vaccine adjuvant that has been safely administered to > 1,400 human subjects.

Fig.1 TLR4 agonists induce Th1 cytokines and type 1 IFN

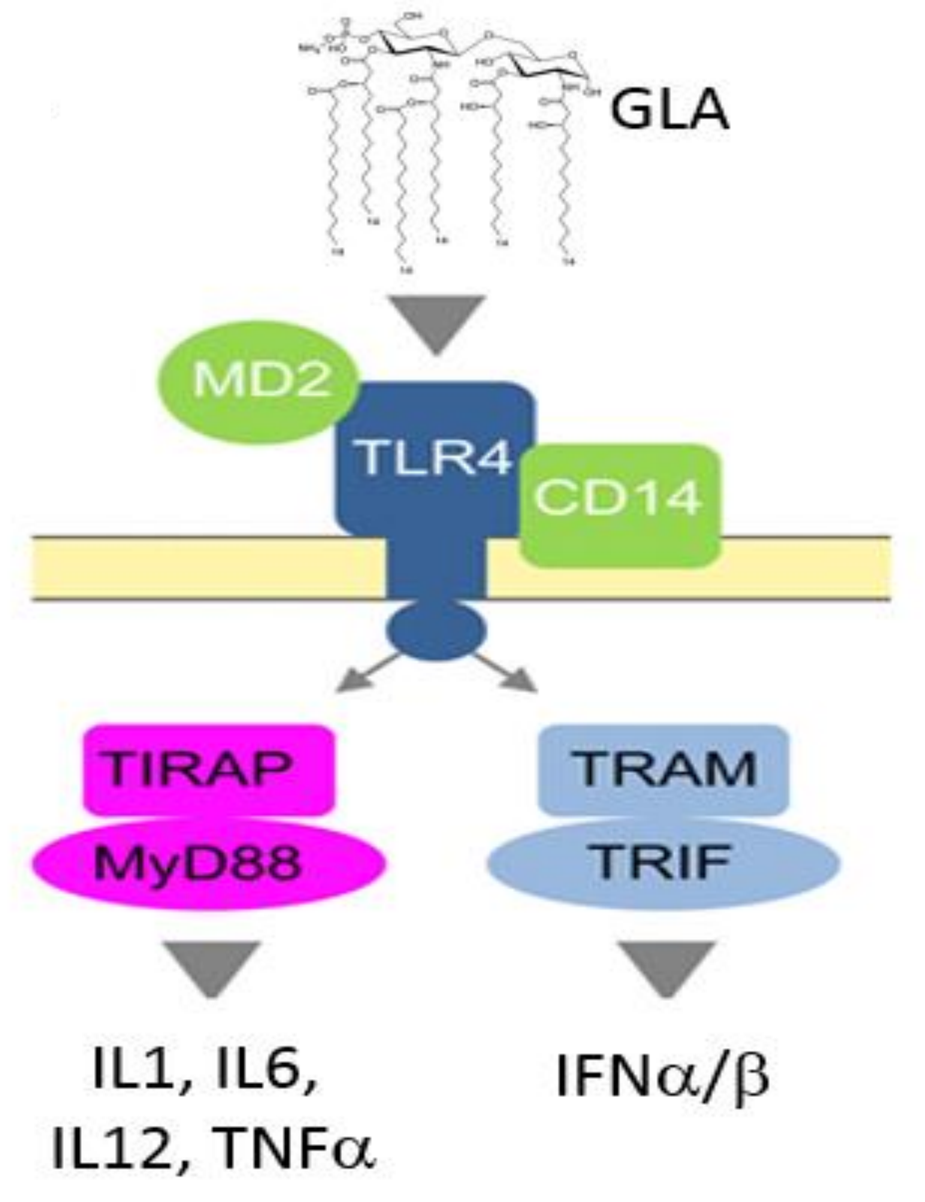
GLA
MD2
TLR4
CD14
TIRAP
MyD88
TRAM
TRIF
IL1, IL6,
IL12, TNFα
IFNα/β

Fig.2 GLA is a potent inducer of innate immune genes

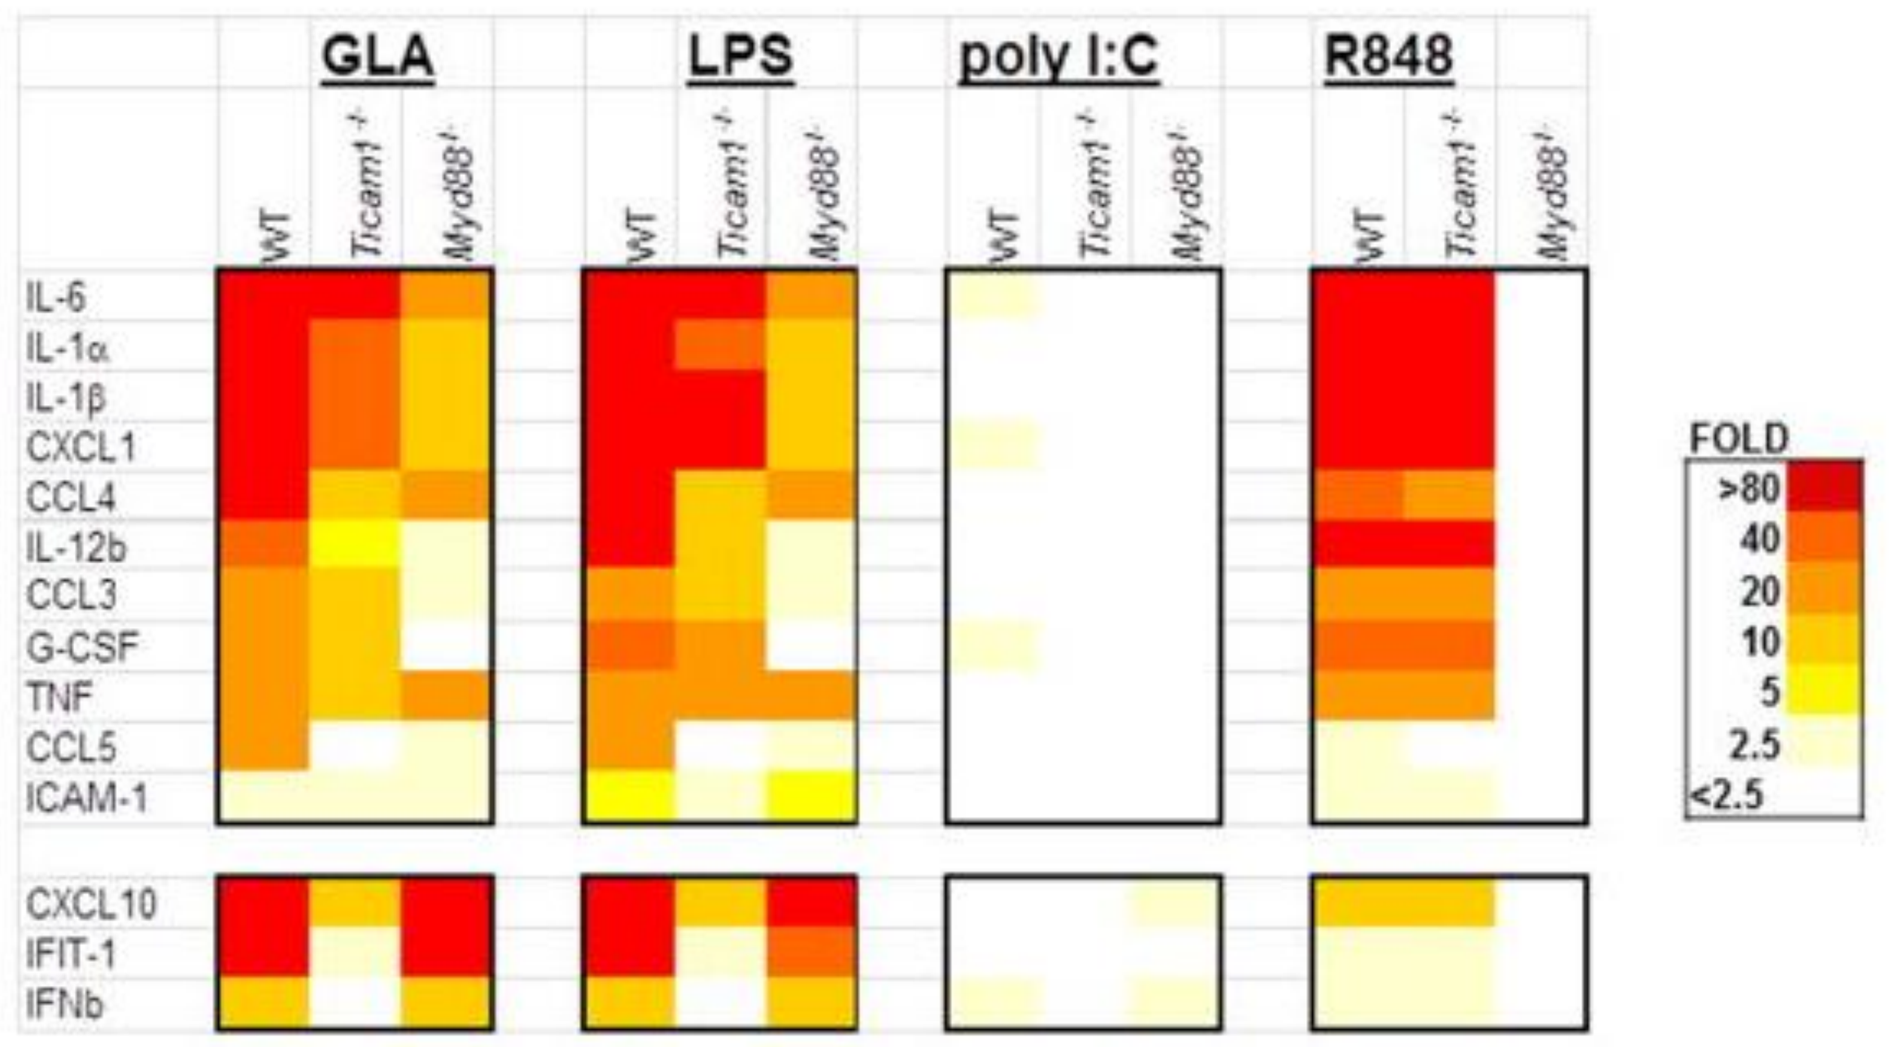
GLA
LPS
poly I:C
R848
WT
Ticam1-/-
Myd88-/-
IL-6
IL-1α
IL-1β
CXCL1
CCL4
IL-12b
CCL3
G-CSF
TNF
CCL5
ICAM-1
CXCL10
IFIT-1
IFNb
FOLD
>80
40
20
10
5
2.5
<2.5

-Orr MT et al, Eur J Immunol, 2013

METHODS

Sample collection: Whole blood was collected from subjects with allergy to Timothy grass pollen under an IRB approved protocol. The allergy to Timothy grass pollen was confirmed by skin prick test or RAST blood test. Peripheral blood mononuclear cells (PBMC) were isolated from whole blood by using Ficoll gradient centrifugation and cells were cryopreserved for batch analysis.

PBMC culture and cytokine measurement: PBMC were cultured with grass pollen *Phleum pretense* extract (10 μg/mL, Greer, Cat # XP28D3A25), with or without GLA (1 μg/mL) for 6 days. Cytokine production was measured by ELISA and Luminex assays using kits from BD Biosciences and Bio-Rad.

IL-12 blockade: Anti-hIL12p70 neutralizing antibody (clone 20C2, BioXcell, Cat#: BE0234) were added to PBMC culture before the addition of *Phl* extract and GLA. The neutralizing antibody was included during the 6 day incubation period.

RESULTS

Fig.3 GLA Decreases the Production of Th2 Cytokines

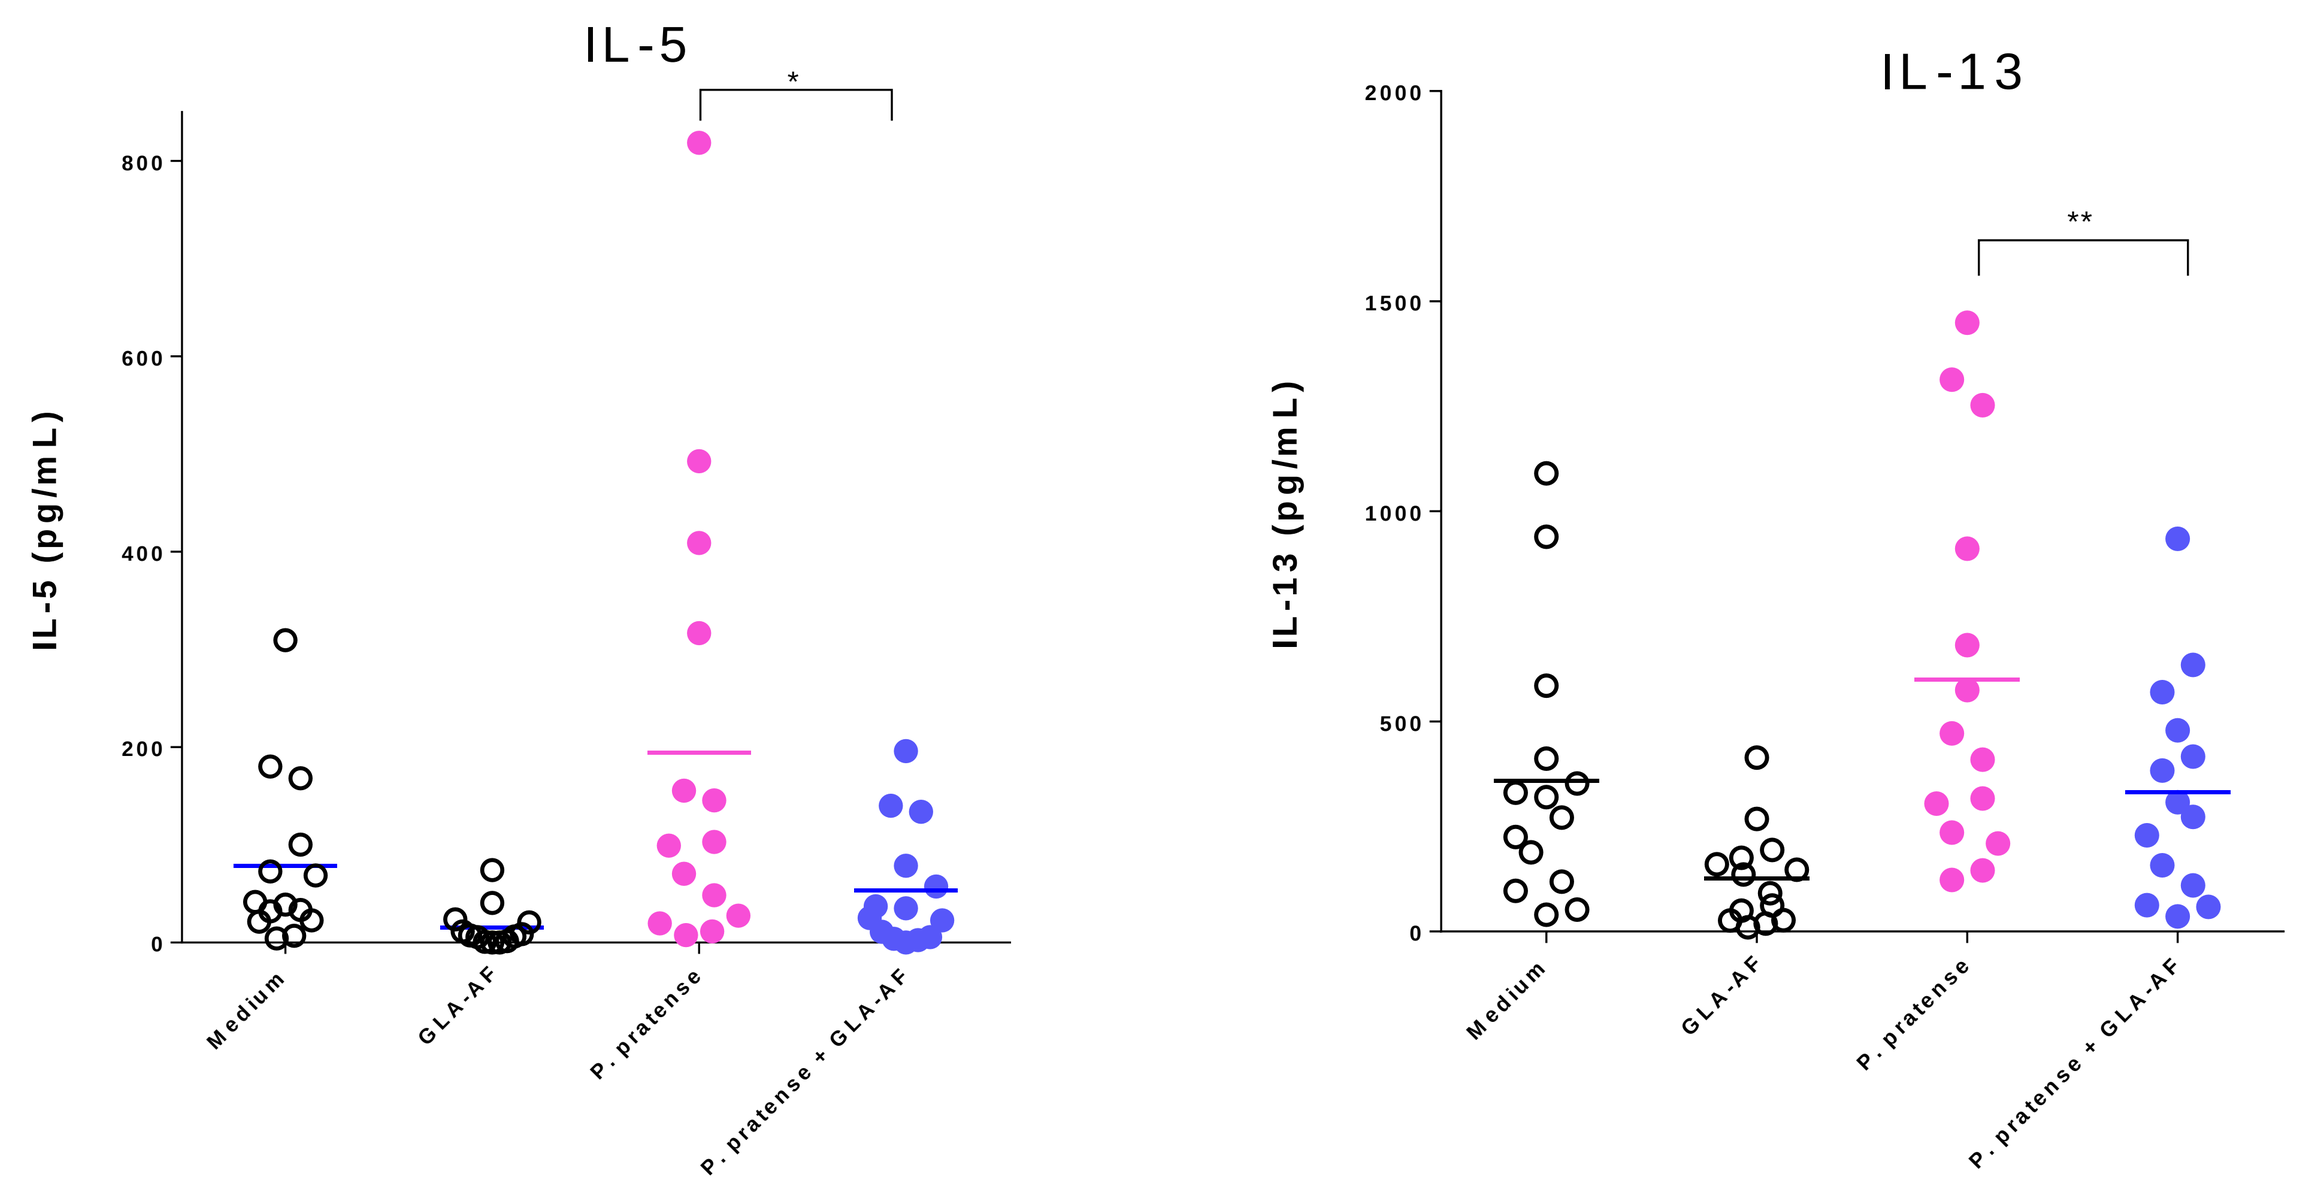
IL-5
IL-5 (pg/mL)
IL-13
IL-13 (pg/mL)
Medium
GLA-AF
P. pratense
P. pratense + GLA-AF
*
**

The levels of Th2 cytokines, IL-5 and IL-13, were significantly decreased in the presence of GLA (1 μg/mL). IL-5 was 213±72 pg/mL in the allergen alone group vs. 60±17 pg/mL in the allergen plus GLA group (p=0.02). IL-13 was 635±140 pg/mL in the allergen alone group vs. 322±75 pg/mL in allergen plus GLA group (p=0.006).

Fig. 4 GLA Increases the Production of Th1 Cytokines

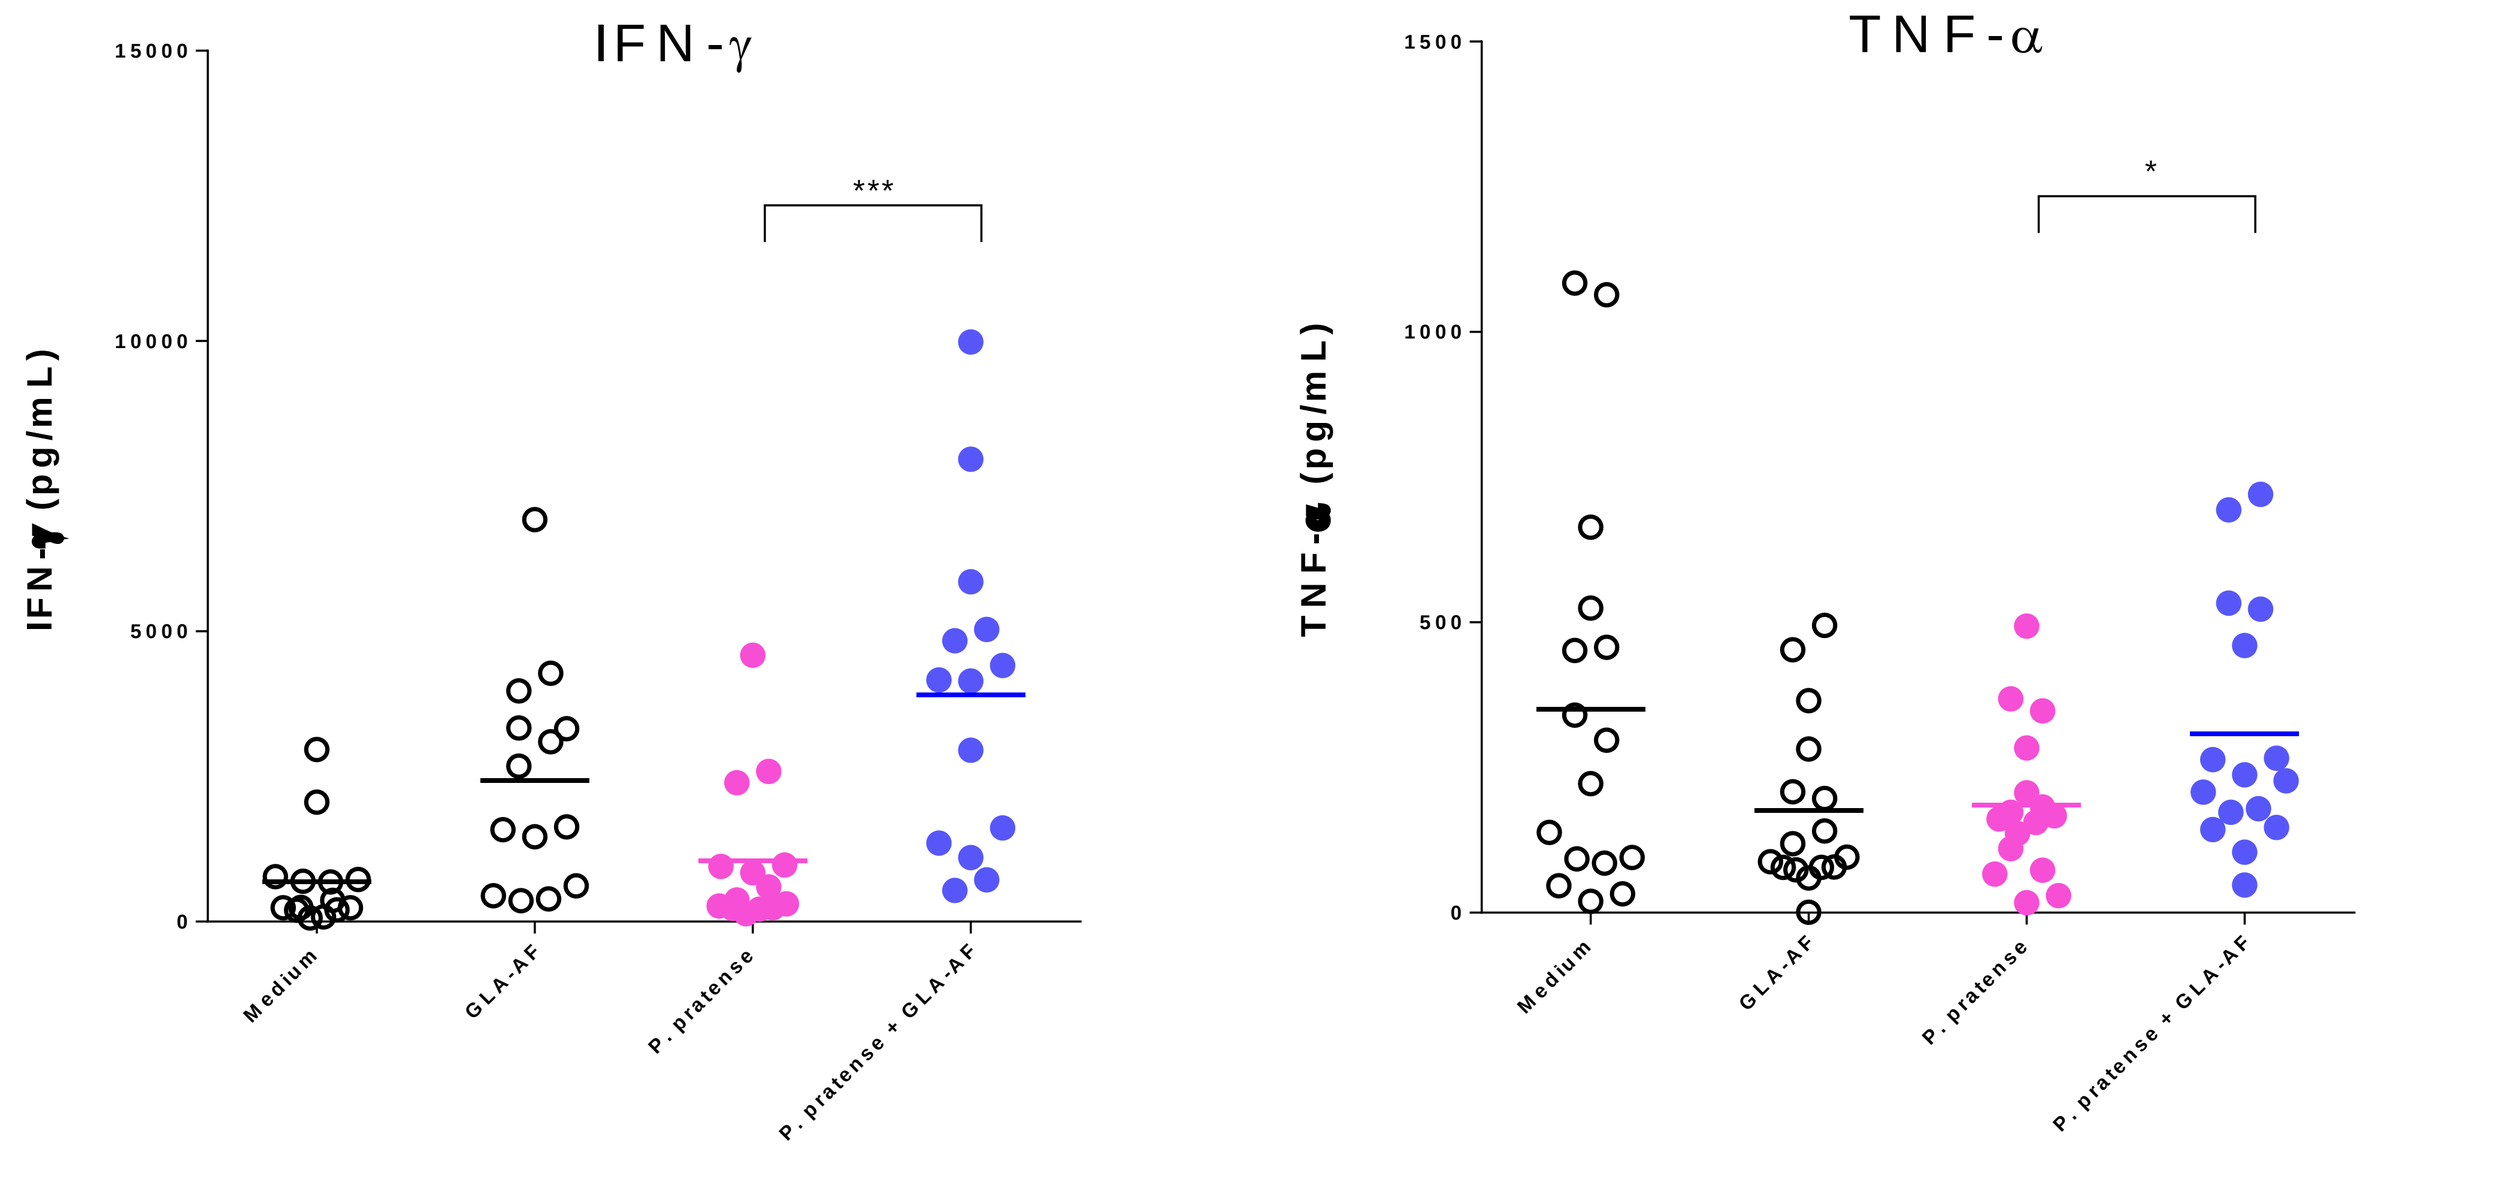
IFN-γ
TNF-α
Medium
GLA-AF
P. pratense
P. pratense + GLA-AF

*

Th1 cytokines, IFNγ and IL-12, were significantly increased in the presence of GLA. IFNγ was 1008±378 pg/mL in the allergen alone group vs. 3842±876 pg/mL in the allergen plus GLA group (p=0.001). TNFα was 186±33 pg/mL in the allergen alone group vs. 308±53 pg/mL in the allergen plus GLA group (p=0.038).

Fig. 5 GLA Induces Th1-polarizing IL-12 and Tolerogenic IL-10

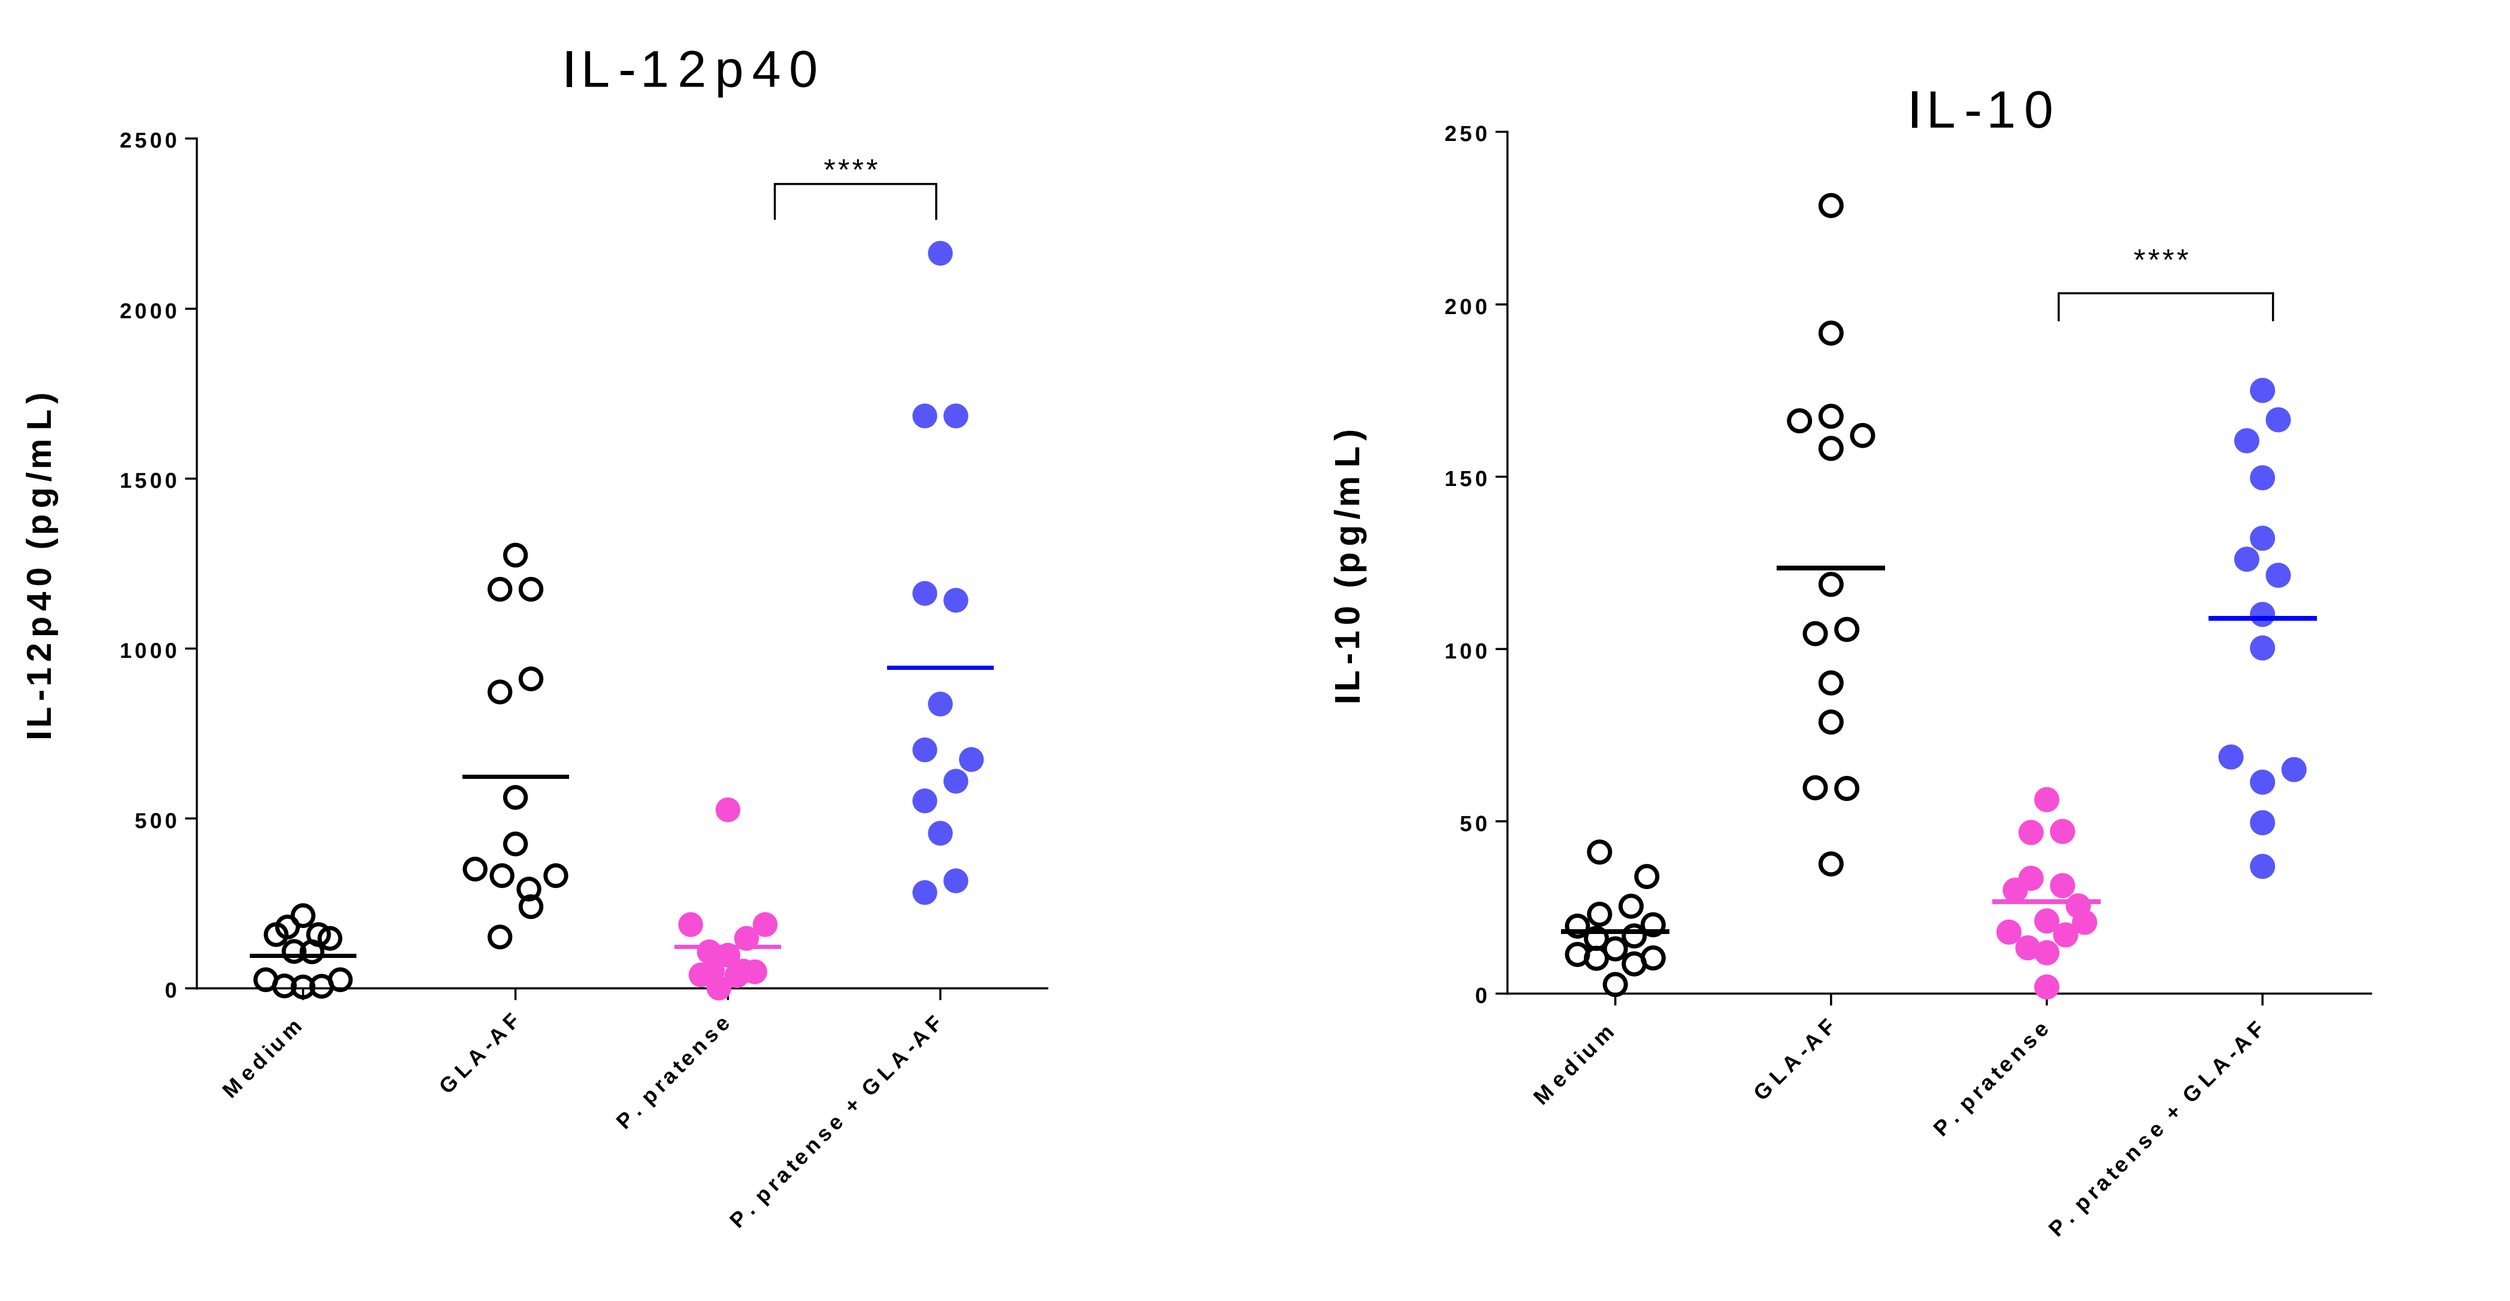
IL-12p40
IL-12p40 (pg/mL)
IL-10
IL-10 (pg/mL)
Medium
GLA-AF
P. pratense
P. pratense + GLA-AF

IL-12p40 was 126±48 pg/mL in allergen alone group vs. 898±179 pg/mL in allergen plus GLA group (p=0.0004). IL-10 was 27±4 pg/mL in allergen alone group vs. 109±12 pg/mL in allergen plus GLA group (p<0.0001).

Fig. 6 Induction of IL-12 and IL-10 by GLA is Dose-dependent

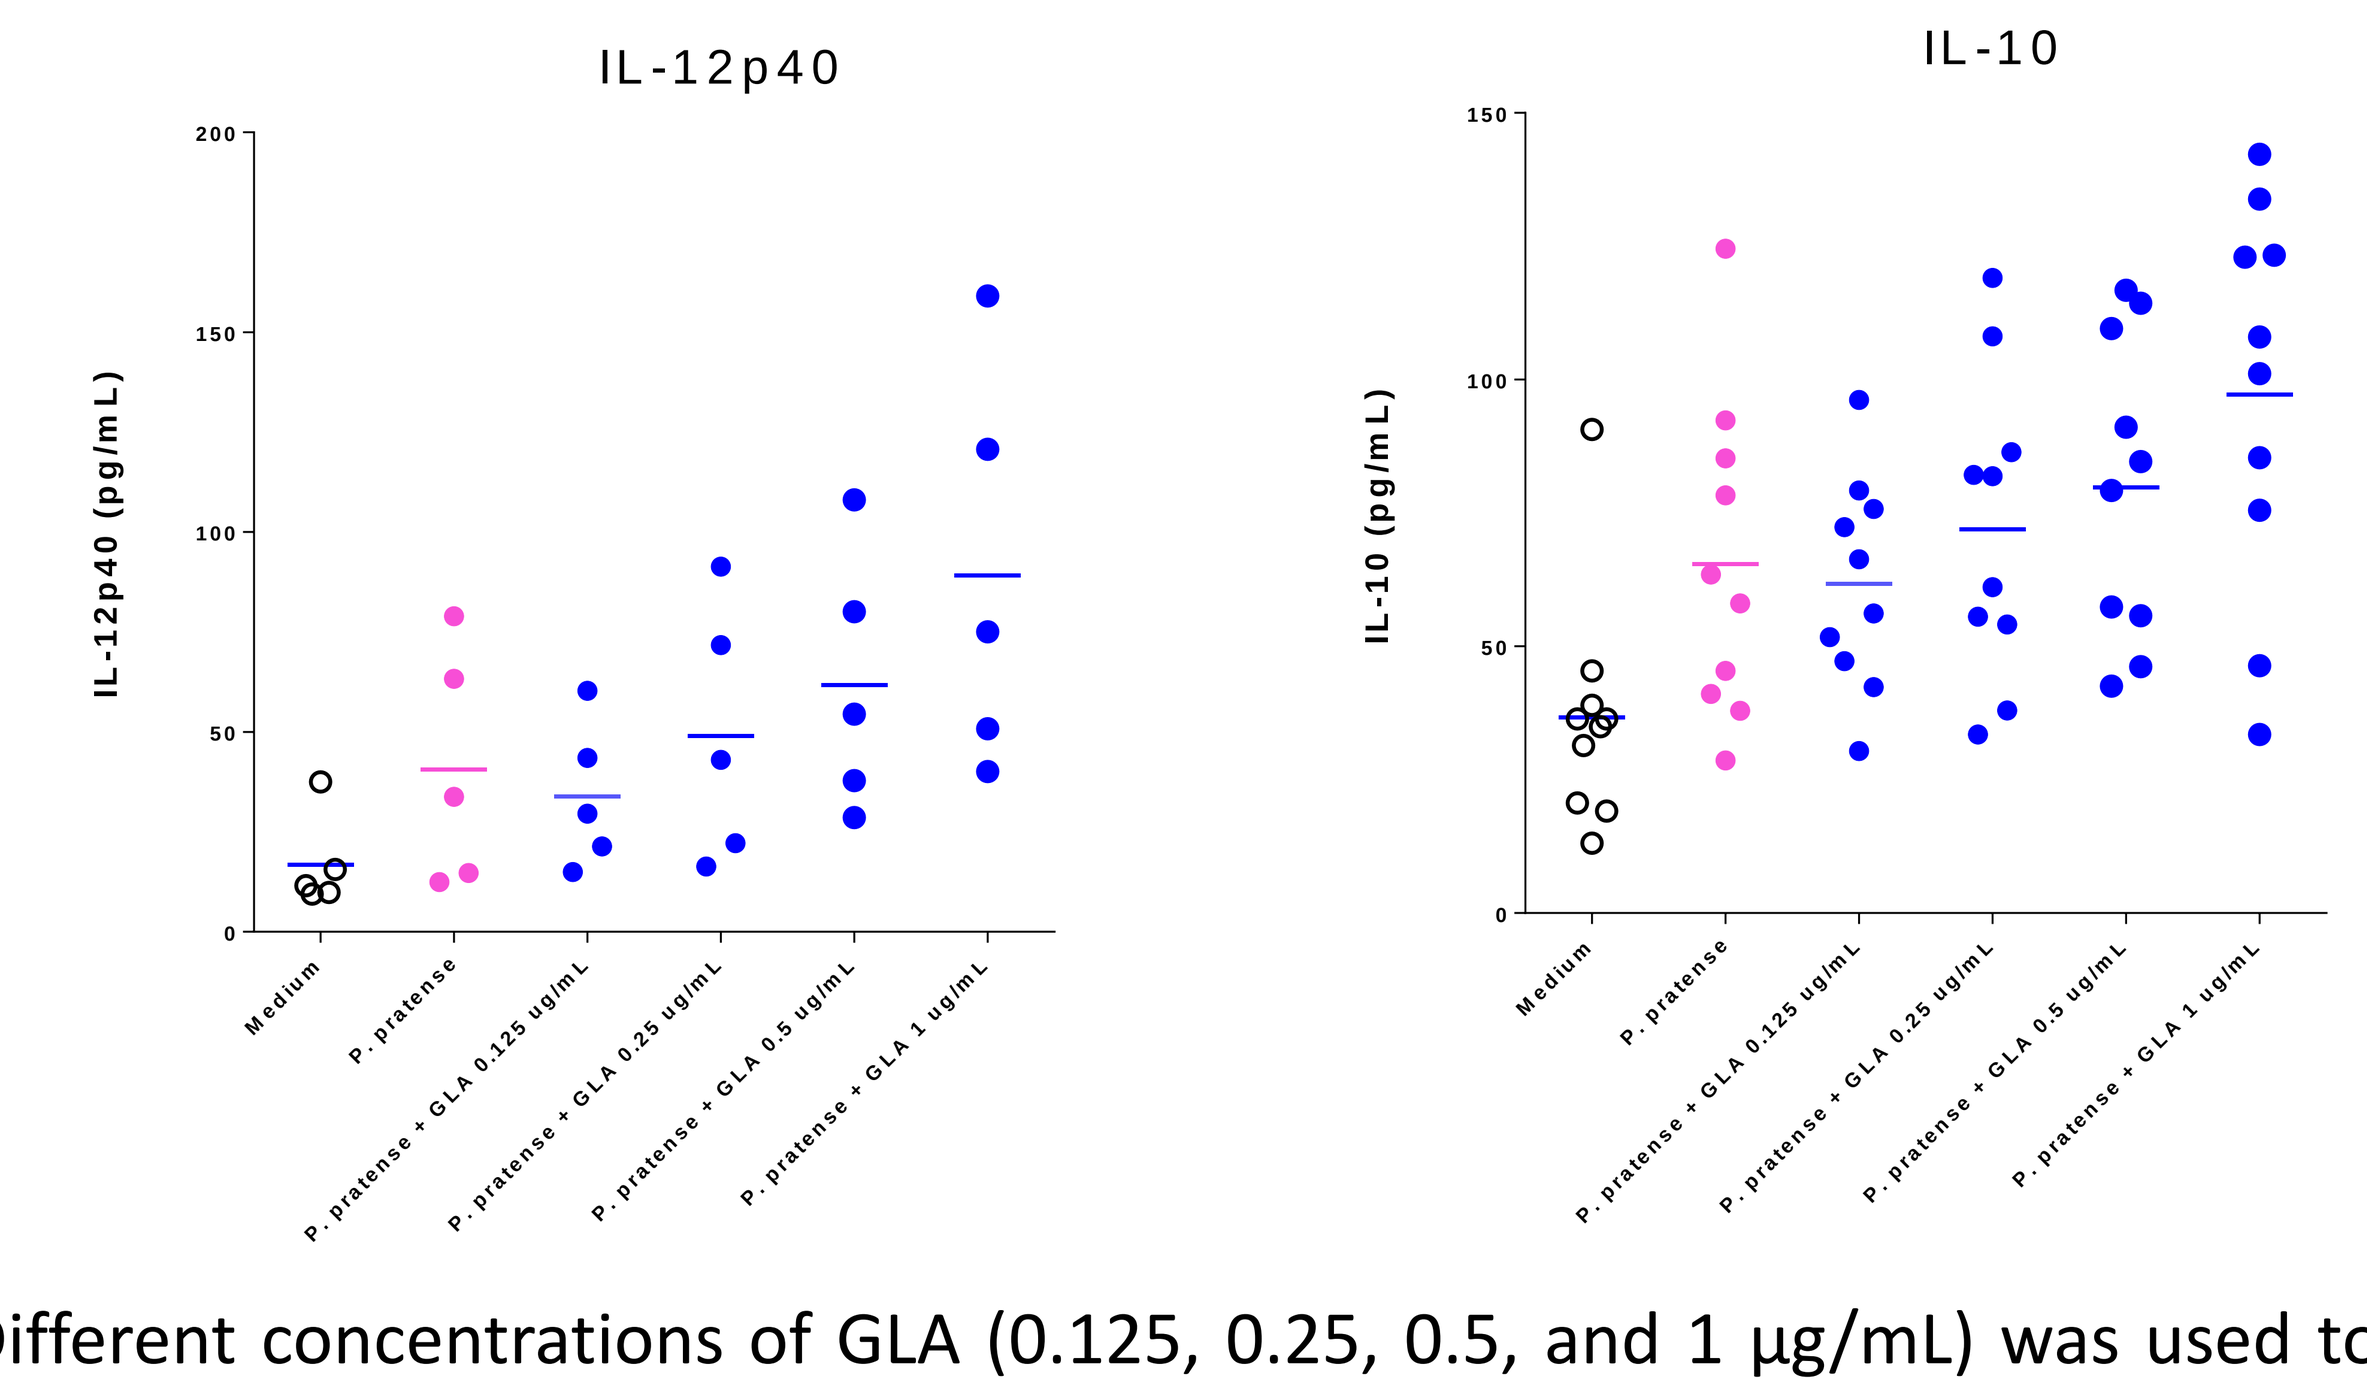
IL-12p40
IL-12p40 (pg/mL)
IL-10
IL-10 (pg/mL)
Medium
P. pratense
P. pratense + GLA 0.125 ug/mL
P. pratense + GLA 0.25 ug/mL
P. pratense + GLA 0.5 ug/mL
P. pratense + GLA 1 ug/mL

Different concentrations of GLA (0.125, 0.25, 0.5, and 1 μg/mL) was used together with *Phl* treatment. Results showed that GLA-mediated induction of IL-12p40 and IL-10 is dose-dependent.

Fig. 7 IL-12 Neutralization Reversed the Effect of GLA

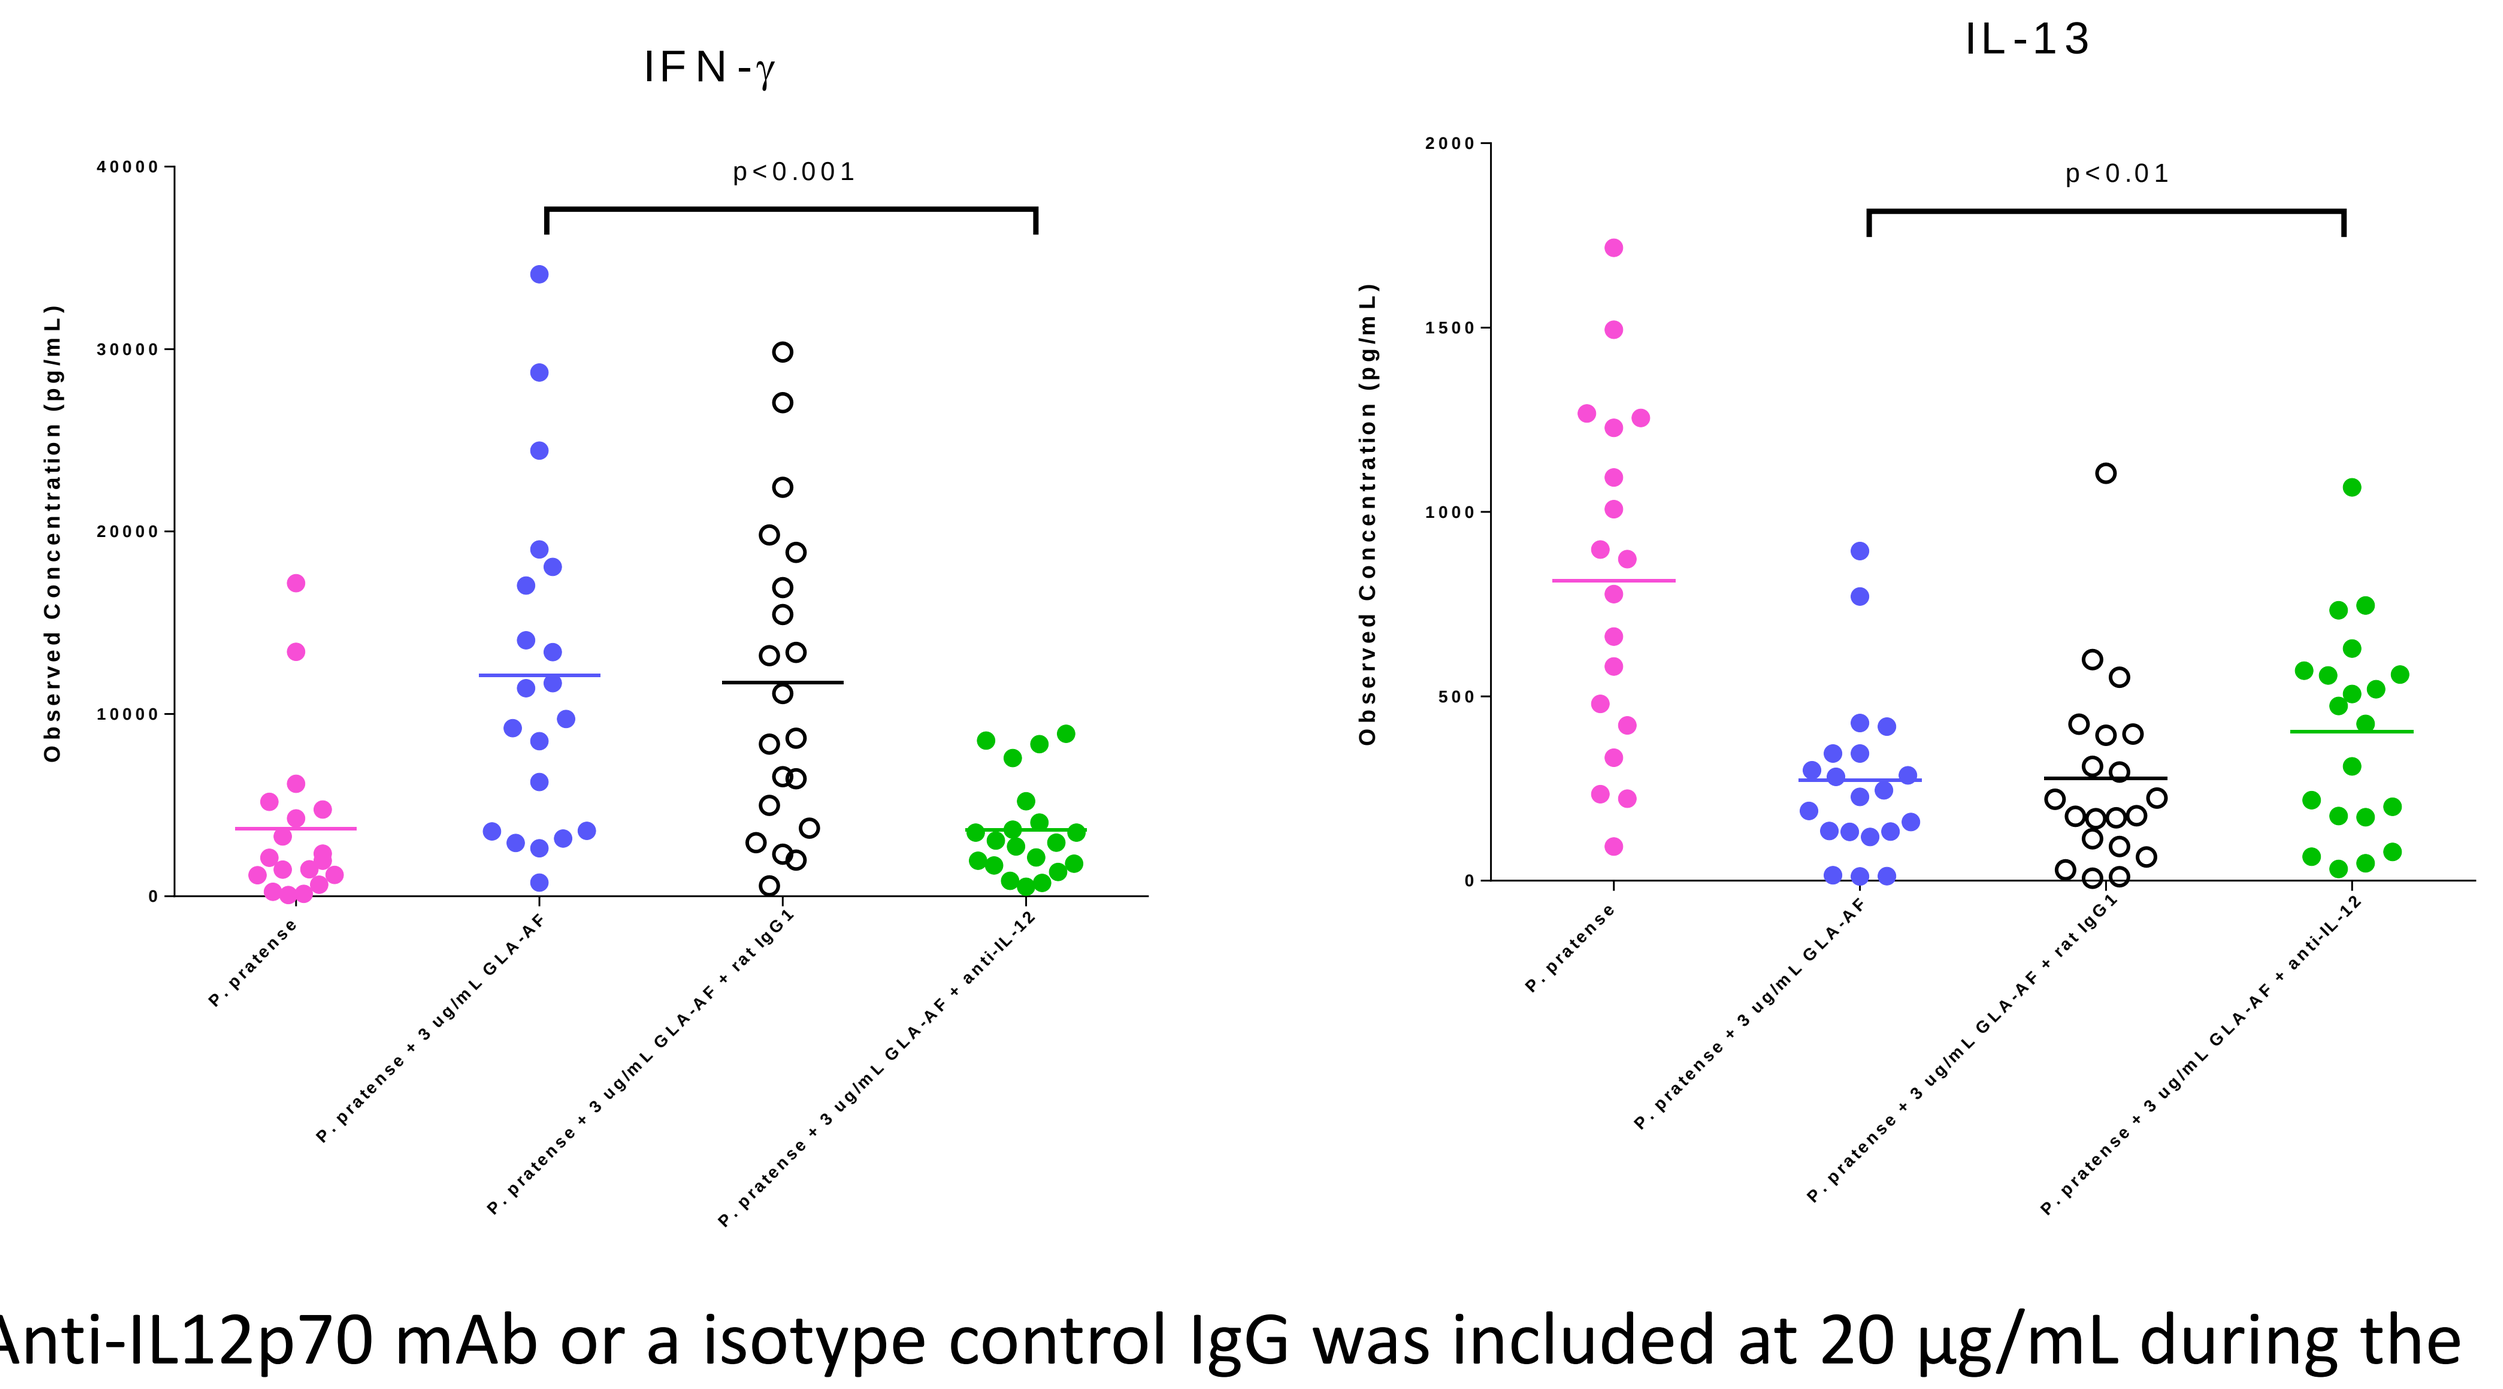
IFN-γ
IL-13
Observed Concentration (pg/mL)
p<0.001
p<0.01
P. pratense
P. pratense + 3 ug/mL GLA-AF
P. pratense + 3 ug/mL GLA-AF + rat IgG1
P. pratense + 3 ug/mL GLA-AF + anti-IL-12

Anti-IL12p70 mAb or a isotype control IgG was included at 20 μg/mL during the treatment with GLA (3 μg/mL). The presence of anti-IL12 but not control IgG inhibited GLA-induced IFNγ significantly (p=0.0002).

SUMMARY

- **GLA modulates the Th1/2 cytokine profiles in allergy patient PBMC by decreasing Th2 cytokines and increasing Th1 cytokines.**
- **The Th1-polarizing effects of GLA in allergy PBMC is dependent on IL-12.**
- **Results from current study suggest GLA may be a promising adjuvant in SIT for pollen allergy that warrants further development.**

Proposed MOA of GLA in Allergy

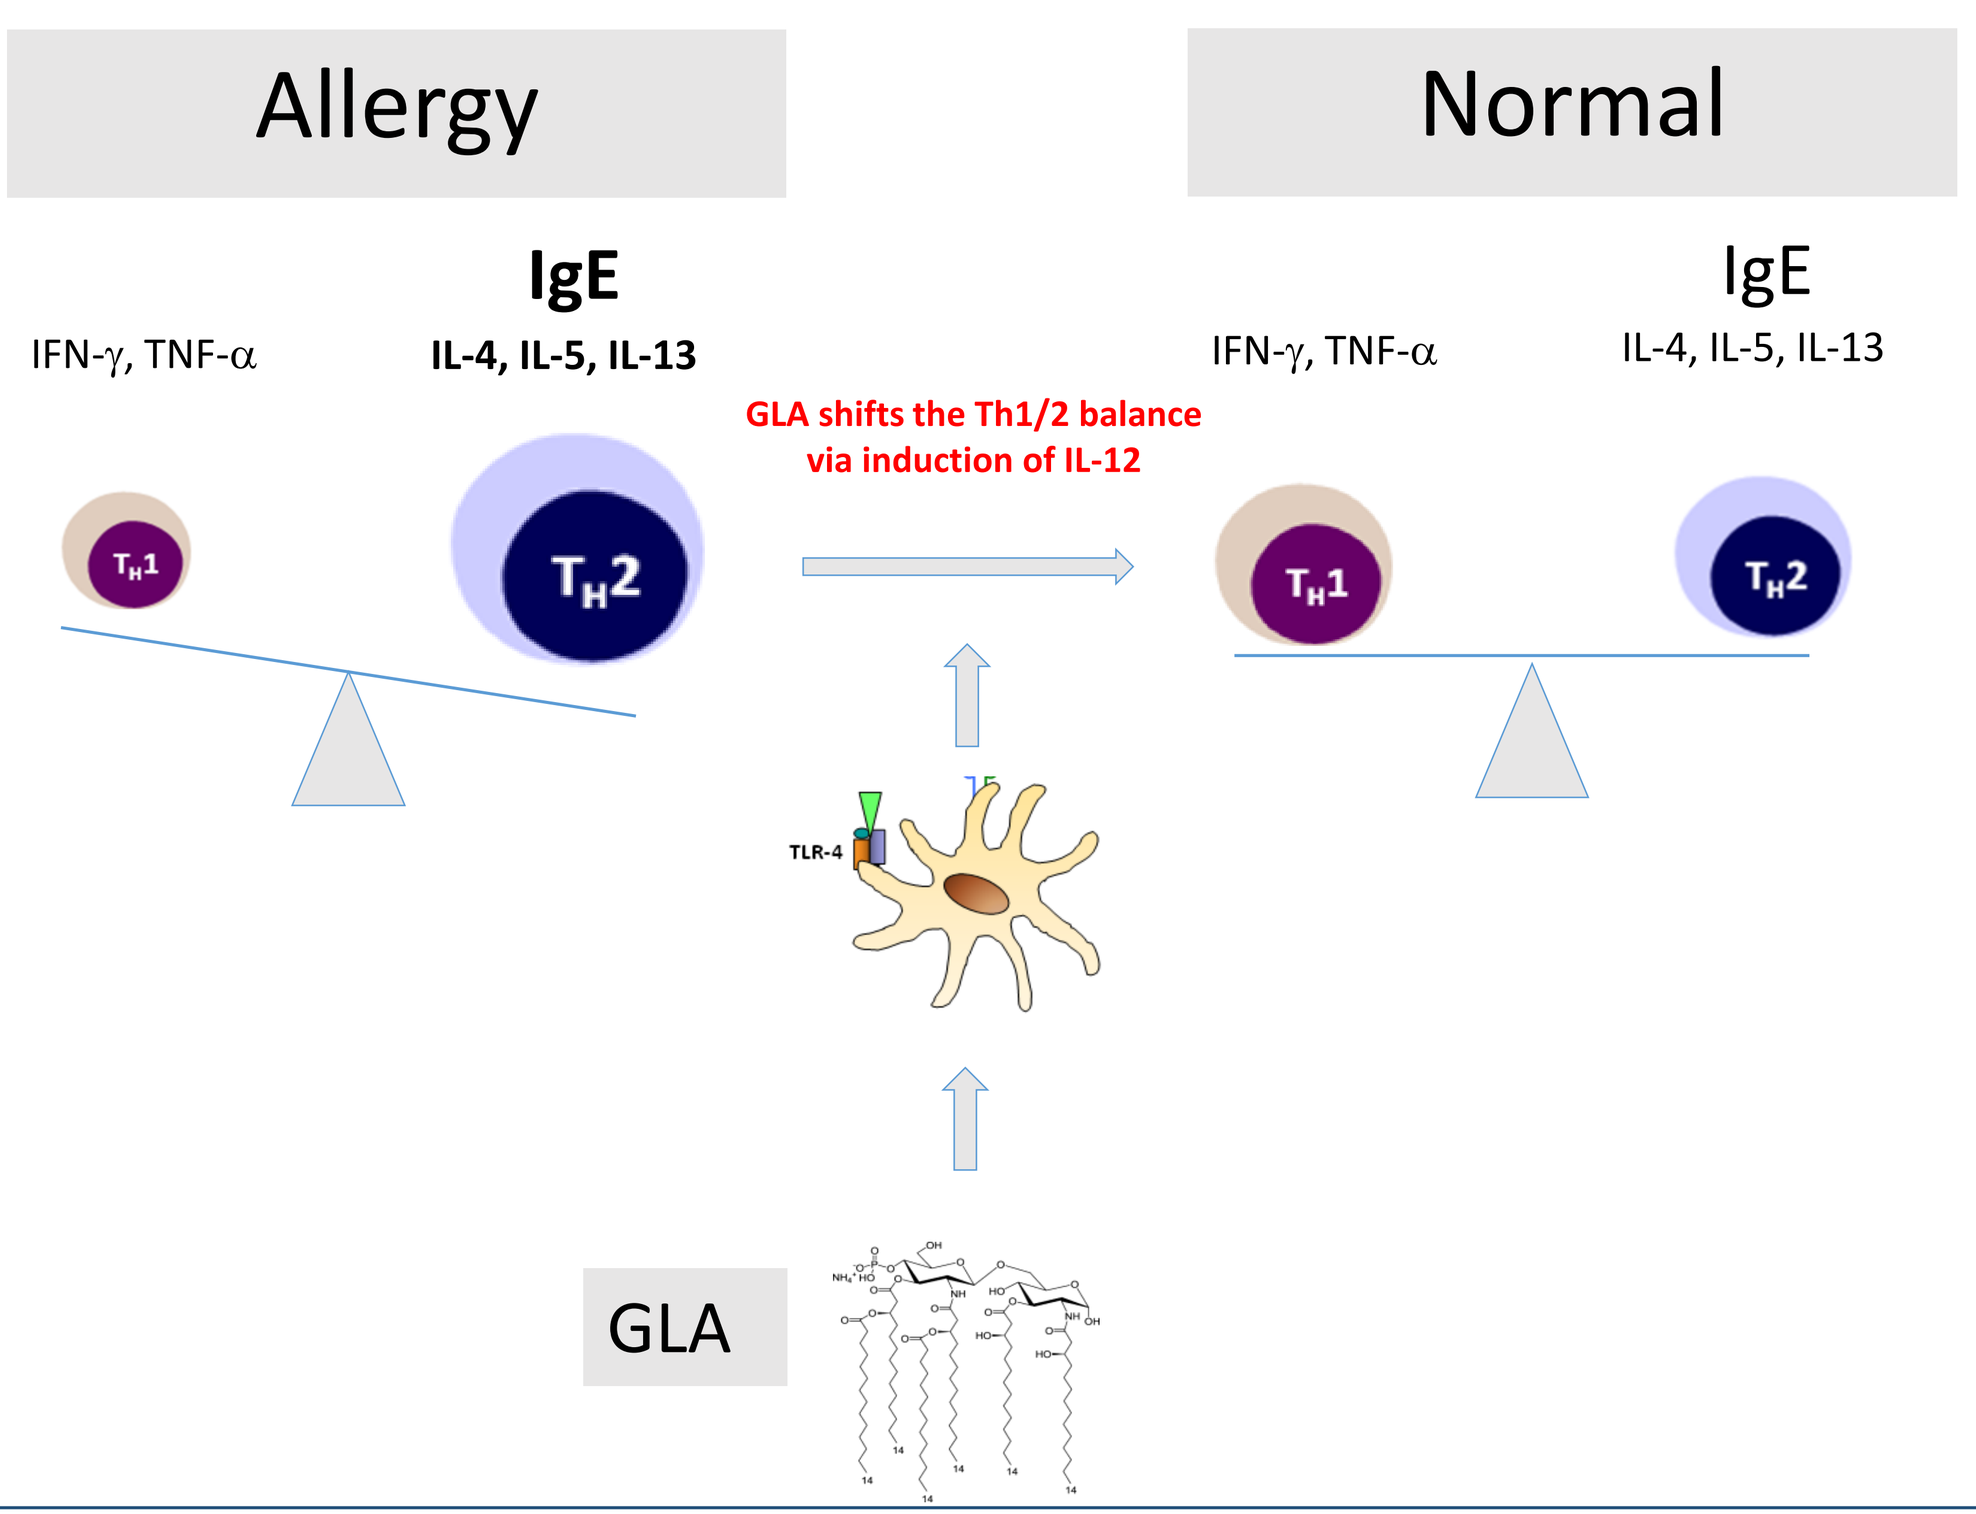
Allergy
Normal
IgE
IFN-γ, TNF-α
IL-4, IL-5, IL-13
IgE
IFN-γ, TNF-α
IL-4, IL-5, IL-13
GLA shifts the Th1/2 balance via induction of IL-12
TH1
TH2
TH1
TH2
TLR-4
GLA

References

1. Development and characterization of synthetic glucopyranosyl lipid adjuvant system as a vaccine adjuvant. PLoS One, 2011 6(1): e16333
2. MyD88 and TRIF synergistic interaction is required for TH1-cell polarization with a synthetic TLR4 agonist adjuvant. Eur J Immunol. 2013 43(9): 2398-408

IMMUNE DESIGN